Serotonin Transporter Occupancy in Raphe Nuclei quantified with PET predicts Treatment Response to Escitalopram and Citalopram in Major Depressive Disorder

MEDICAL UNIVERSITY OF VIENNA

Akimova Elena[1], Lanzenberger Rupert[1], Savli Markus[1], Häusler Daniela[2], Wadsak Wolfgang[2], Spindelegger Christoph[2], Moser Ulrike[2], Fink Martin[2], Mitterhauser Markus[2], Kletter Kurt[2], Kasper Siegfried[1] *

[1] *Department of Psychiatry and Psychotherapy, Medical University of Vienna, Austria*
[2] *Department of Nuclear Medicine, Medical University of Vienna, Austria*

biol-psychiatry@meduniwien.ac.at

CLINICAL DIVISION OF BIOLOGICAL PSYCHIATRY

Background:

Impaired serotonin neurotransmission has been implicated in the pathophysiology of affective disorders based on the results of numerous preclinical and clinical investigations. Interest is now focusing on the underlying mechanisms involved in the effect of serotonin reuptake inhibitors (SSRIs), currently the most frequently prescribed and effective medication in the treatment of depression. Evidence points to the key role of the serotonin transporter (SERT) found in its highest concentration in the raphe nuclei, as SSRIs bind to SERT thereby blocking the reuptake of serotonin from the synaptic cleft (1). The predictive value of SERT availability by SSRIs in the clinical response to antidepressants has recently become an interesting and controversial topic in the treatment of major depressive disorder (MDD) (2,3). Our longitudinal study with 3 PET scans tests the hypothesis that serotonin transporter availability and occupancy in patients with MDD may be biological markers predicting treatment outcome.

Methods:

The study was designed as a double-blind, randomized, longitudinal PET study with 2 parallel treatment groups. Patients with MDD were treated with equivalent doses of 10mg S-Citalopram (10mg/day Escitalopram (50% of subjects) or 20mg/day Citalopram). Using PET and the highly selective radioligand [^{11}C]DASB, we evaluated SERT binding before treatment (1st PET) in 18 patients, after a single oral dose (2nd PET) and after a treatment period of 3 weeks (3rd PET). SERT availability at baseline and SSRI treatment induced occupancies of SERT after a single dose and after a treatment period of 3 weeks were quantified in ten brain regions. SERT availability was correlated with treatment response assessed by the Hamilton Depression Rating Scale (HAM-D) after 3 weeks of treatment *(see fig2)*.

Results:

There was no significant interaction between S-Citalopram plasma levels and treatment response, neither in the case of single- nor prolonged treatment. Lower pretreatment SERT availability in raphe regions and midbrain indicated improved treatment response 3 weeks later. We focused our analyses on 10 brain regions in which we expected high to medium SERT availability, and a reliable binding measurement. Among the regions investigated, the pre-treatment SERT availability significantly explained the variability in the proportional decrease of HAM-D scores in the midbrain and raphe regions only. Significant correlations (Spearman) between the decrease in HAM-D scores and SERT availability were found in the midbrain (rho=-0.643, p=0.004), mesial raphe nucleus (MRN) (rho=-0.672, p=0.002), and dorsal raphe nucleus (rho=-0.583, p=0.011, not surviving correction for multiple testing) *(see fig1a)*. A significant relationship between treatment response and transporter occupancy (2nd PET) was observed in MRN (rho=-0.577, p=0.012) *(see fig1b)*.

References:

1 *Meyer JH. Imaging the serotonin transporter during major depre sive disorder and antidepressant treatment. J Psychiatry Neurosci 2007; 32(2): 86-102.*

2 *Kugaya A, Sanacora G, Staley JK, Malison RT, Bozkurt A et al. Brain serotonin transporter availability predicts treatment response to selective serotonin reuptake inhibitors. Biol Psychiatry 2004; 56(7): 497-502.*

3 *Lundberg J, Christophersen JS, Petersen KB, Loft H, Halldin C, Farde L. PET measurement of serotonin transporter occupancy: a comparison of escitalopram and citalopram. The International Journal of Neuropsychopharmacology 2007; 10(06): 777-785.*

Acknowledgements:

This work was supported by an investigator-initiated and unrestricted research grant from H. Lundbeck A/S, Denmark to S. Kasper. The study protocol has been planned by the authors who retained full academic control and are publishing the study independently.

CONCLUSION:

To our knowledge this is the first PET study with [^{11}C] DASB demonstrating a significant relationship between treatment response and SERT availability and occupancy. Our results show that lower availability of SERT before treatment in the midbrain and raphe regions may indicate better treatment outcome in response to SSRIs. With our study we have attempted to provide further insight into the controversial subject of biological markers and their role as predictive instruments, contributing to an improved clinical management of MDD.

fig1

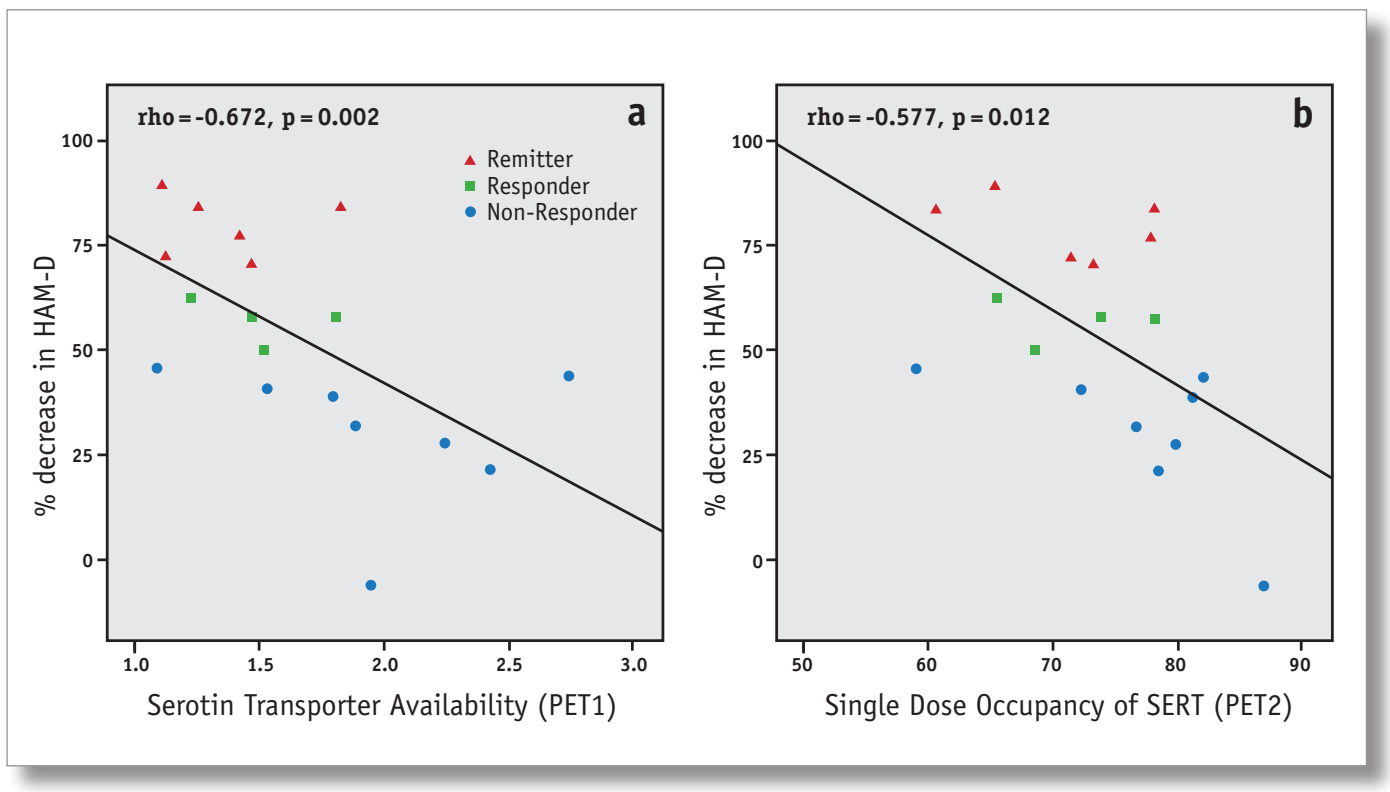
rho = -0.672, p = 0.002
a
Remitter
Responder
Non-Responder
% decrease in HAM-D
100
75
50
25
0
1.0
1.5
2.0
2.5
3.0
Serotin Transporter Availability (PET1)
rho = -0.577, p = 0.012
b
% decrease in HAM-D
50
60
70
80
90
Single Dose Occupancy of SERT (PET2)

Correlations between the proportional decrease of depression (Hamilton Depression Rating Scale (HAM-D) scores) and baseline (1st PET) serotonin transporter (SERT) availability (a) and single dose occupancy (b) in the mesial raphe nucleus by the enantiomer S-Citalopram. Green squares indicate responders, who showed a 50% or greater reduction relative to initial scores, blue circles are non responders (below 50% reduction). All responders displaying a final score (3rd PET) of ≤ 7 are marked as red triangles (remitters). Lines represent the linear fit.

fig2

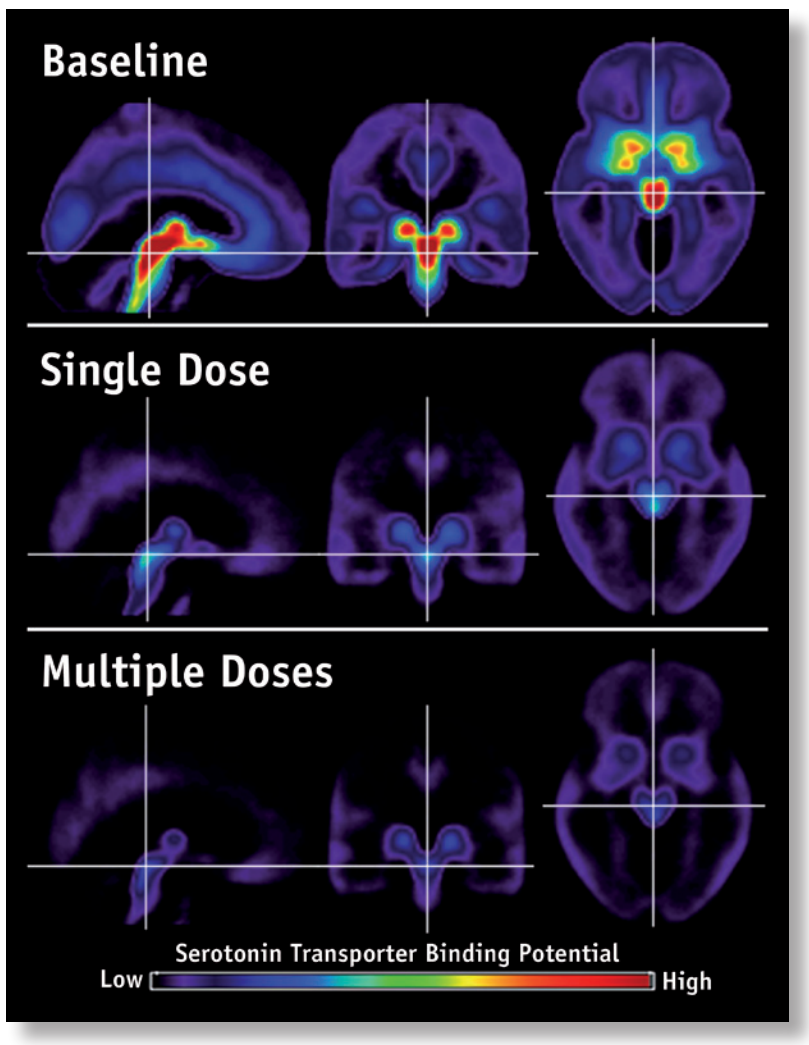
Baseline
Single Dose
Multiple Doses
Serotonin Transporter Binding Potential
Low
High

Mean [^{11}C]DASB binding under baseline, single dose and multiple doses conditions (triplanar PET images). The parametric maps correspond to individual summed, averaged and normalized scans and represent the mean serotonin transporter distribution in depressed patients before and after SSRIs intake (10 mg/day of the enantiomer S-Citalopram). A single dose led to a considerable transporter blockage resulting in a mean occupancy of 74.8% ± 6.8%. Three weeks of SSRIs treatment raised occupancies to 84.0% ± 7.0%.

Conflict of interest and financial disclosures:

S. Kasper has received grants/research support from Eli Lilly, Lundbeck, Bristol-Myers Squibb, Servier, Sepracor, GlaxoSmithKline, Organon; has served as a consultant or on advisory boards for AstraZeneca, Austrian Science Fund, Bristol-Myers Squibb, GlaxoSmithKline, Eli Lilly, Lundbeck, Pfizer, Organon, Sepracor, Janssen, and Novartis; and has served on speakers' bureaus for AstraZeneca, Eli Lilly, Lundbeck, Servier, Sepracor and Janssen. R. Lanzenberger has received a travel grant and research support from Lundbeck A/S, furthermore conference speaker honoraria from Lundbeck and AstraZeneca. C. Spindelegger has received a travel grant from Lundbeck; U. Moser has received travel grants from Bristol-Myers Squibb and Astra Zeneca.